Immunohistochemical Characterization of Kidney Lesions in Cynomolgus Monkeys using a Triple Staining Method

Sally Price[1], Dan Heathcote[1], Samantha Steele[1], Elmarie Bodes[2], Mausumee Guha[3], Margareta Bielenstein[4], Robert G. Caccese[5], Tom Simpson[6], David B. Stong[3], Annabelle Heier[1]

AstraZeneca [1]Pathology Department, Safety Assessment Alderley Park, UK; [2]Emerging Product Team, Neuroscience Therapy Area, [5]Safety Pharmacology, [3]Safety Assessment, [6]Medicinal Chemistry, Wilmington, USA; [4]Clinical Pharmacology & DMPK, Södertälje, Sweden

INTRODUCTION

Most studies investigating segment specific nephrotoxicity have been carried out in rodents (Gautier et al, 2010, Hoffmann et al, 2010). However, cynomolgus monkeys are frequently used as a non rodent species for safety studies and often display similar pathology. A major problem with investigating nephrotoxicity in the monkey and identifying potential biomarkers is that very few antibodies cross react with monkey tissues. A new test compound was given to cynomolgus monkeys for up to one month. The kidney was identified as main target organ. By histological evaluation of H&E slides, the tubular lesions could not be attributed to a particular nephron segment. In order to better characterize the lesions and identify a potential urinary biomarker, the aim was to localize the lesions to particular nephron segments using immnohistochemistry (IHC) methods. Markers investigated were calbindin D28K, aquaporin 1 and aquaporin 2. Calbindin D28K is a vitamin D-dependent calcium binding protein expressed in the distal tubule and collecting ducts (Bindels et al, 1991). Urinary levels of this marker are correlated with damage to renal distal tubules in patients following cisplatin chemotherapy (Takashi et al, 1996). Aquaporin 1 is a water channel expressed specifically in proximal tubules and thin limb of the loop of Henle. Urinary aquaporin 2 is a membrane bound water channel found specifically in principal cells of the collecting ducts in the kidney (Nielsen et al, 1993).

RESULTS

Microscopic evaluation of the kidneys from animals dosed 50 mg/kg/day for 4 or 5 days showed multifocal severe degeneration and necrosis of tubuli, extending from the outer cortex to the inner medulla. Affected tubuli were markedly dilated and filled with neutrophils, debris and protein-rich material (Fig. 1A, B and C). Dosing at 25 mg/kg/day for 7 days led to prominent simple dilatation of tubuli (Fig. 1D) and multifocal tubular degeneration. Longer dosing (20 mg/kg/day for up to 1 month) led to multifocal degeneration and regeneration in parallel (Fig. 1E). Multiple tubuli were lined by degenerated epithelium with desquamated epithelial cells, neutrophil casts or increased proteinaceous material in the lumen. Tubular degeneration was associated with a mixed inflammatory cell infiltration of neutrophils, and fewer lymphocytes and macrophages in the surrounding interstitium. Tubular regeneration varied from increased basophilia of the tubular epithelium with flattened to low cuboidal cells to hyperplastic foci of tubular epithelium with piling up of nuclei. While the lesions affected mainly the tubuli and the adjacent interstitium, the glomeruli appeared morphologically normal (Fig. 2A and B).

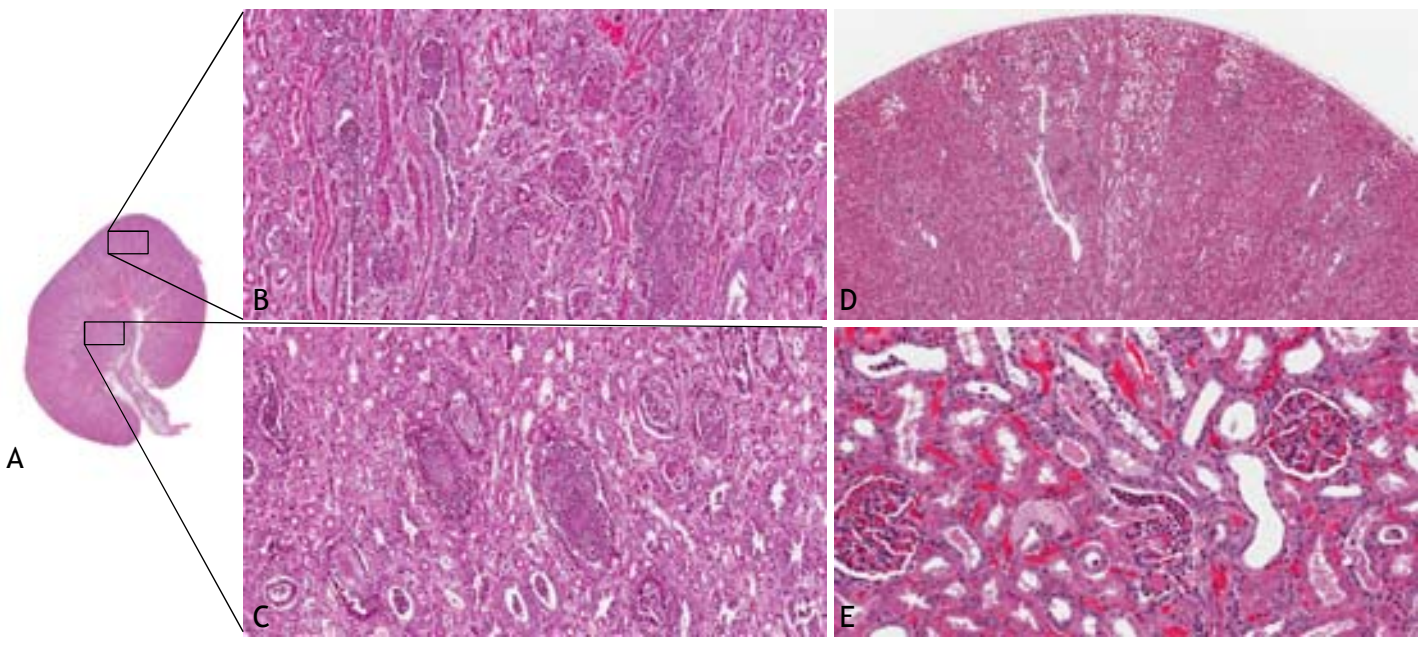
A
B
C
D
E

Figure 1: Kidney, cynomolgus monkey, H&E staining. **A** Female dosed with 50 mg/kg/day for 5 days, x3, **B** x100, **C** x100. **D** Male dosed with 25 mg/kg/day for 7 days, x15. **E** Male dosed with 20 mg/kg/day for 17 days, x200.

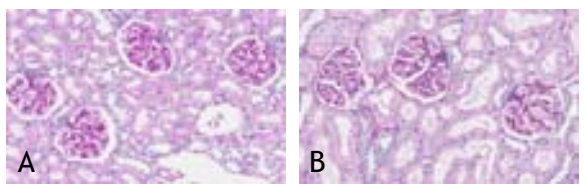
A
B

Figure 2: Kidney, female monkey, PAS staining showing that glomeruli appeared morphologically unaffected.
A Control **B** Animal dosed with 20 mg/kg/day for 1 month, x200.

The most notable finding in serum chemistry was an increase in serum urea (+284%) in the female dosed at 10 mg/kg/day for 7 days (Table 1). In animals dosed with 20 mg/kg/day for 1 month, there was a slight increase in mean urea concentration at week 2 and 4, which returned to normal levels at the end of the recovery period. No significant effects on urinary parameters were noted at the end of the 1-month dosing.

Table 1: Clinical chemistry findings after 7 daily doses *

Dose level (mg/kg/day)	10		25	
	M	F	M	F
Urea	-	+284%	+49%	+75%
Creatinine	-	+85%	+34%	+56%

Table 2: Clinical chemistry findings in the 1-month study with 1 month recovery *

Dose level (mg/kg/day)	5		10		20	
Urea	M	F	M	F	M	F
Week 2	-	-	+33%	+71%	+50%	+58%
Week 4	-	-	+51%	+55%	+35%	+48%
Week 8	ND	ND	ND	ND	-	-

* Difference (%) compared to control and pretest values; - no change; ND = not done

MATERIAL AND METHODS

The in-life part of the animal study was conducted at Ricerca Biosciences (formerly MDS Pharma Services), Lyon, France in general compliance with ICH guidelines. Cynomolgus monkeys (*Macaca fascicularis*) were administered the test compound via naso-gastric intubation. In two separate studies, animals were given daily doses of 10, 25 and 50 mg/kg/day for up to 7 days or daily doses of 5, 10 and 20 mg/kg/day for 1 month. Dosing with 20 mg/kg/day was followed by a 1-month dosing free period. For the 7-day study, blood samples for assessment of serum biochemistry were collected once pretest and on day 7. For the 1-month study, blood and urine were sampled twice pretest and in study week 2, 4 and 8. All animals were necropsied, tissue samples were fixed in 10% neutral formalin for routine processing (for 2-3 weeks) and paraffin-embedded. Slides were cut at nominal 5 µm and stained with hematoxylin and eosin (H&E) and with Periodic Acid Schiff (PAS).
IHC was performed as described previously (Price et al, 2010). For the triple stain, rabbit polyclonal anti-aquaporin 2 (Sigma, Saint Louis, Missouri, USA) was the first primary antibody to be applied (1:4000, 30 min) and detected using rabbit-specific EnVision®+ System-HRP polymer kit (Dako, UK), with diaminobenzidine (DAB) as the chromogen (10 min). Mouse monoclonal anti-calbindin D28K antibody (1:100, 30 min, Sigma) was the next antibody to be applied. This was detected using the mouse-specific EnVision®+ System-HRP (Dako, UK), polymer kit (Dako) with Vector SG (Vector Laboratories) as the chromogen (10 min). Rabbit anti-rat aquaporin 1 (Alpha Diagnostic International, San Antonio, Texas, USA) was the final antibody to be applied (1:200, 60 min) and this was detected using a rabbit/mouse polymer alkaline phosphatase kit (Dako, 30 min) with permanent red (Dako) as the chromogen (10 min). In between each primary antibody, sections were incubated in serum free block (Dako) for 15 min. For single stains, anti-calbindin D28K was applied at 1:1000 for 30 min and anti-aquaporin 2 was applied at 1:4000 for 60 min. Dual Link HRP Labelled Polymer (DAKO) was then applied to slides treated with the un-conjugated antibodies for 30 min (mouse or rabbit, respectively) and the immunoreactivity was visualised using DAB.
An aliquot of urine from multiple time points during the 1-month study (pretest and weeks 2, 4 and 8) was used to identify potential biomarkers. Human KidneyMAP™ v. 1.0 panel was used (Rules Based Medicine (RBM), TX). The markers that were cross reactive with the human panel were ß2-microglobulin, calbindin, clusterin, CTGF, cystatin C, NGAL, THP, timp 1 and VEGF. Urinary biomarker data was assessed by analysis of variance (ANOVA) followed by a Tukey's post hoc test.

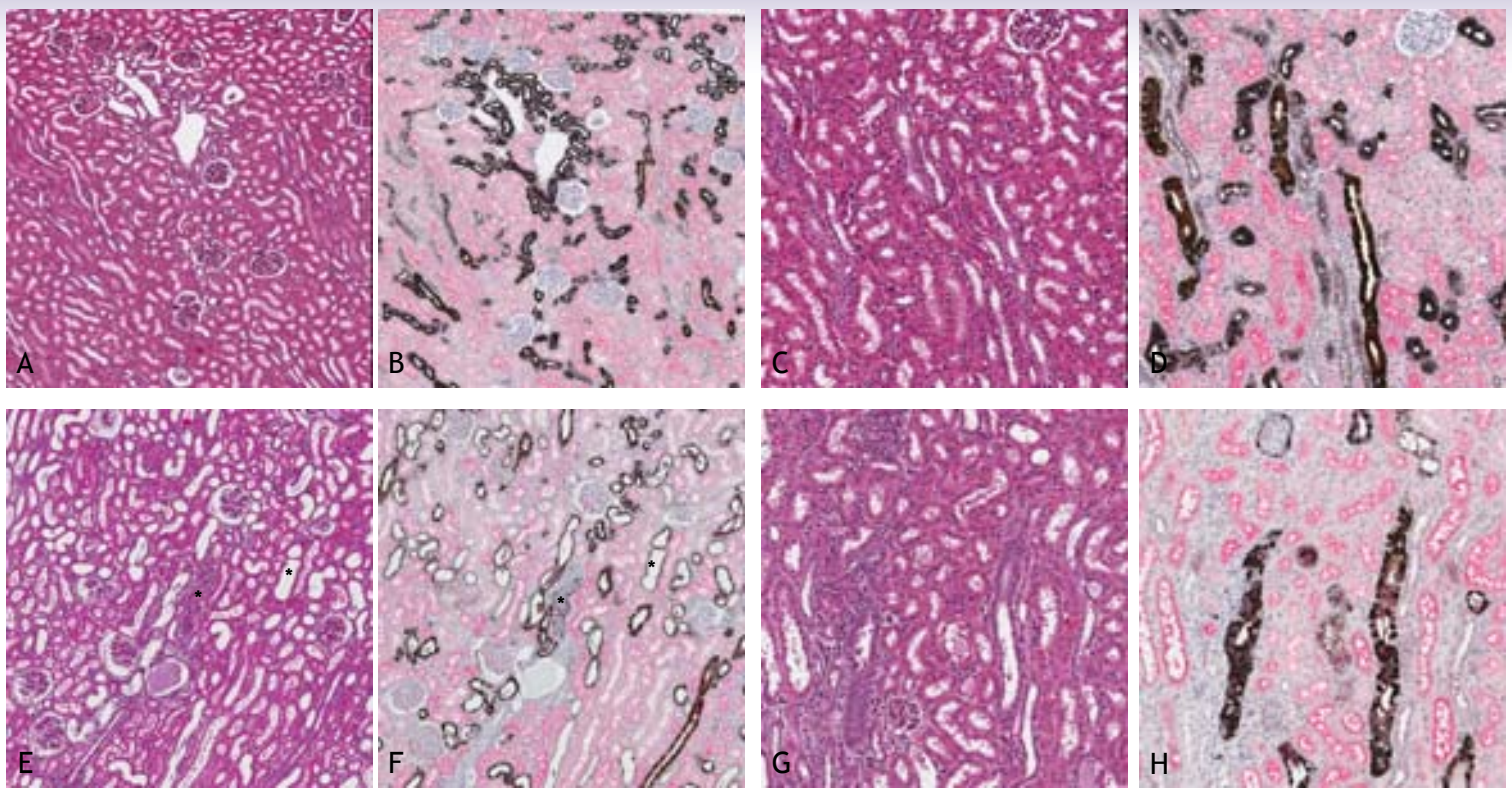
A
B
C
D
E
F
G
H

Figure 3: Serial sections of monkey kidney were compared by H&E (**A, C, E, G**) and by the triple IHC method (**B, D, F, H**). Proximal tubuli were stained with aquaporin 1 (red), distal tubuli with calbindin D28K (dark blue) and collecting ducts with both aquaporin 2 (brown) and calbindin D28K.
A, B Female control, x50. **C, D** Female control monkey, x100. **E, F** Female monkey dosed with 25 mg/kg/day for 7 days, x50. **G, H** Male monkey dosed with 20 mg/kg/day for 1 month, x100.

A triple stain IHC method was developed (Fig. 3A and B), using antibodies against aquaporin 1 (proximal tubule), aquaporin 2 (collecting ducts) and calbindin D28K (distal tubules and collecting ducts). Tubuli presenting with degeneration, neutrophil casts or dilatation by H&E, stained positive for calbindin D28K and aquaporin 2 (* Fig. 3E and F). In comparison to control monkeys (Fig. 3C), an increase in tubular basophilia, interpreted as regeneration, was present in animals dosed with 5, 10 or 20 mg/kg/day for 1 month, often associated with desquamation of tubular epithelial cells and peritubular inflammatory cells (Fig. 3G). Affected tubuli could be identified as distal tubules and collecting ducts by the triple stain (Fig. 3H).
A single IHC stain for calbindin D28K stained all tubuli presenting with dilatation or degeneration positive (Fig. 4A). A few of these affected tubuli also stained positive with the single IHC stain for aquaporin 2 (Fig. 4B).

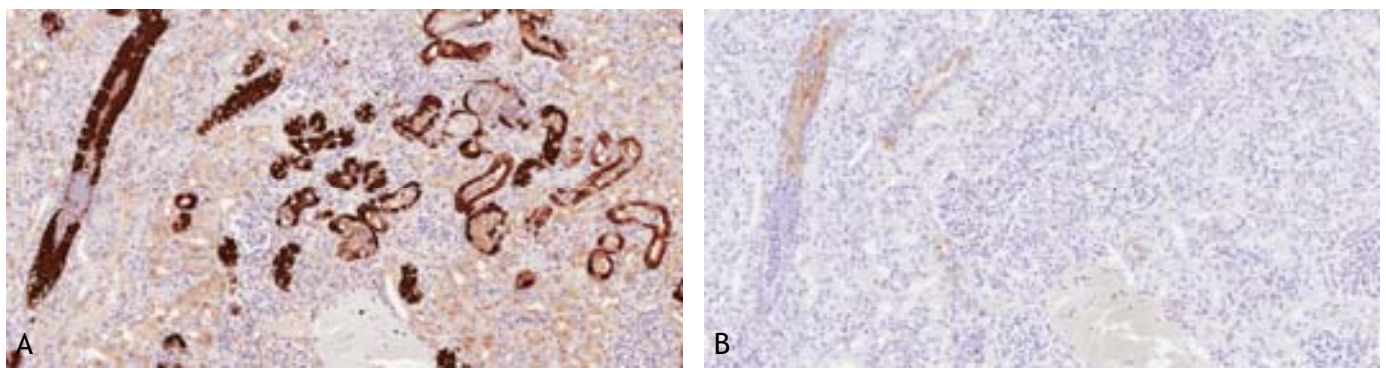
A
B

Figure 4: Serial kidney sections of a male cynomolgus monkey dosed with 50 kg/kg/day for 4 days.
A IHC for calbindin D28K, staining tubuli dilated by neutrophils and debris positive, identifying them as distal tubules or collecting ducts, x100. **B** IHC for aquaporin2, staining few affected tubuli, confirming them as collecting ducts, x100.

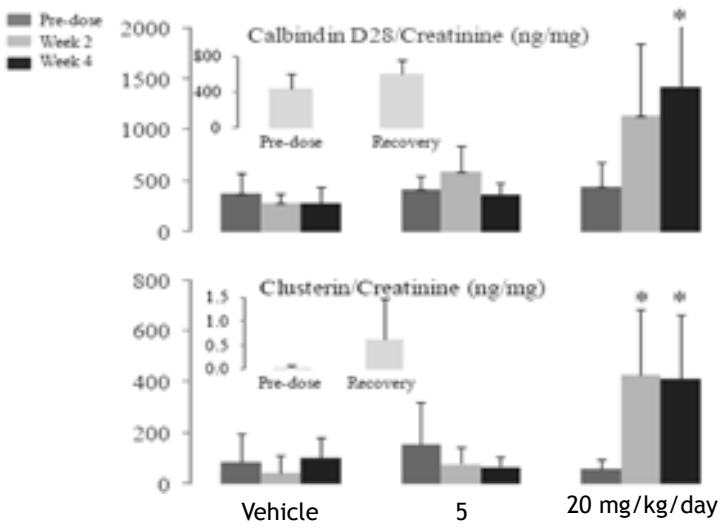
Pre-dose
Week 2
Week 4
Calbindin D28/Creatinine (ng/mg)
Pre-dose
Recovery
Clusterin/Creatinine (ng/mg)
Pre-dose
Recovery
Vehicle
5
20 mg/kg/day

Figure 5: Urinary biomarker results after dosing for 1 month.

Urine creatinine was unchanged during the study and therefore served as an appropriate normalization marker for the segment specific biomarkers. During the 1-month dosing, the test compound consistently increased urinary levels of calbindin D28K and clusterin (a marker of generalized kidney injury in multiple species) in animals dosed with 20 mg/kg/day. An increase was noted when compared to vehicle dosed controls as well as pre-dose levels. Increases were found in both the absolute levels and also when the data was normalized to urinary creatinine (Fig. 5). No significant changes were observed for ß2-microglobulin, CTGF, cystatin C, NGAL, THP, timp 1 and VEGF.

DISCUSSION

The IHC method developed was successfully used to label proximal tubules (aquaporin 1), distal tubules (calbindin D28K) and collecting ducts (aquaporin 2 and calbindin D28K) in the kidney of the cynomologous monkey. By histological evaluation of H&E slides, the tubular lesions caused by the test compound could not be attributed to a particular nephron segment. Urea and creatinine were only slightly increased in the majority of animals and are known to be insensitive markers for kidney injury (Schnellmann 2008). Using the triple stain IHC method, the tubular lesions could be located to the distal tubules and collecting ducts. These IHC findings correlated with increased urinary excretion of calbindin D28K. The calbindin D28K antibody shows good species cross reactivity (Sourial et al, 2009), suggesting that calbindin D28K could be a useful urinary biomarker for indicating nephrotoxicity caused by this compound and others affecting the distal nephron.
In summary, using a triple IHC stain for aquaporin 1, aquaporin 2 and calbindin D28K we could localize tubular lesions induced by the test compound to the distal nephron and correlate them with a urinary biomarker with potential utility in the clinic.

ACKNOWLEDGEMENTS

We would like to thank Ricerca Biosciences (formerly MDS Pharma Services) for the excellent collaboration and Kevin Randall for his outstanding technical support.

References

Bindels RJ, Hartog A, Timmermans JA, van Os CH. Immunocytochemical localization of calbindin-D28k, calbindin-D9k and parvalbumin in rat kidney. Contrib Nephrol. 1991;91:7-13.
Gautier J-C, Riefke B, Walter J, Kurth P, Mylecraine, Guilpin V, Barlow N, Gury T, Hoffman D, Ennulat D, Schuster K, Harpur E, Petit S. Evaluation of biomarkers of nephrotoxicity in two strains of rat treated with cisplatin. Tox Pathol 2010. August 17 [Epub ahead of print].
Hoffmann D, Adler M, Vaidya VS, Rached E, Mulrane L, Gallagher WM, Callanan JJ, Gautier JC, Matheis K, Staedtler F, Dieterle F, Brandenburg A, Sposny A, Hewitt P, Ellinger-Ziegelbauer H, Bonventre JV, Dekant W, Mally A. Performance of novel kidney biomarkers in preclinical toxicity studies. Toxicol Sci 2010;116(1):8-22.
Nielsen S, DiGiovanni SR, Christensen EI, Knepper MA, Harris HW. Cellular and subcellular immunolocalization of vasopressin-regulated water channel in rat kidney. Proc Natl Acad Sci USA. 1993;90(24):11663-7.
Price SA, Davies D, Rowlinson R, Copley CG, Roche A, Falkenberg FW, Riccardi D, Betton GR. Characterization of renal papillary antigen 1 (RPA-1), a biomarker of renal papillary necrosis. Toxicol Pathol. 2010;38(3):346-58.
Schnellmann RG In: Casarett and Doull's Toxicology: The Basic Science of Poisons. CD Klaasen 2008 7th ed. McGraw-Hill, New York, p 583.
Sourial S, Searchfield L, Schuppe-Koistinen I, Betton GR, Riccardi D, Price SA. Application of Dolichos biflorus in immunoassay detection of kidney collecting duct biomarkers. Biomarkers. 2010;15(5):424-35.
Takashi M, Zhu Y, Miyake K, Kato K. Urinary 28-kD calbindin-D as a new marker for damage to distal renal tubules caused by cisplatin-based chemotherapy. Urol Int. 1996;56(3):174-9.

AstraZeneca